Poster number: 146

Pharmacokinetic-Pharmacodynamic Modeling of Switching From Aripiprazole Monohydrate to TV-46000, a Long-Acting Subcutaneous Risperidone, in a Virtual Population of Patients With Schizophrenia

Itay Perlstein,[1] Jonathan Meyer,[2] Sharath Kumar,[3] Vijay Ivaturi,[3,4] Nikita Ramwani,[3] Pavan Rathina Muthu Kumar,[3] Kelli R. Franzenburg,[5] Mark Suett,[6] Rolf Hansen,[7] Avia Merenlender Wagner,[8] Arti Phatak,[7] Rajendra Singh[5]

[1]Magic Wand Research LLC, Philadelphia, PA, United States; [2]University of California, Department of Psychiatry, San Diego, CA, United States; [3]Pumas-AI Inc, Dover, DE, United States; [4]Manipal College of Pharmaceutical Sciences, Manipal Academy of Higher Education, Center for Pharmacometrics, Manipal, Karnataka, India; [5]Teva Branded Pharmaceutical Products R&D LLC, West Chester, PA, United States; [6]Teva UK Limited, Global Medical Affairs, Harlow, United Kingdom; [7]Teva Branded Pharmaceutical Products R&D LLC, Parsippany, NJ, United States; [8]Teva Pharmaceutical Industries Ltd., Netanya, Israel

Objective: To simulate switching from aripiprazole monohydrate to TV-46000 and estimate dopamine D2-receptor occupancy and antagonism using population pharmacokinetic-pharmacodynamic modeling

Background

- Differences in pharmacological properties between various long-acting injectable antipsychotics (LAIs), combined with a lack of clinical studies on transitioning patients between them, contribute to barriers during switching related to patient acceptance, clinical practice, and knowledge[1]
- TV-46000 (UZEDY®) is a subcutaneous risperidone LAI formulation administered once monthly (q1m) or once every 2 months (q2m) approved by the United States Food and Drug Administration for the treatment of schizophrenia in adults[2]
 - Prior to initiating TV-46000, tolerability with oral risperidone should be established[2]
- Transitioning from aripiprazole monohydrate once monthly (Abilify Maintena®, AOM), a partial dopamine D2-receptor (D2R) agonist (which produces approximately 75% antagonism), to TV-46000, a D2R antagonist, requires consideration of pharmacodynamic and pharmacokinetic differences
- Although both agents bind D2R, aripiprazole—due to its higher binding affinity and partial intrinsic activity—exhibits greater D2R occupancy (D2RO) to achieve comparable antagonism. In contrast, risperidone, a more potent D2R antagonist, may exert comparable or stronger antagonistic effects even at lower levels of D2RO[3–4]
 - Although antipsychotics bind a variety of different receptors, therapeutic efficacy among D2R blockers is mainly thought to be related to D2RO and antagonism[5]
- Data on how D2RO and antagonism are impacted by switching may support clinicians who want to transition from AOM to TV-46000

Methods

- Published population pharmacokinetic models were used to simulate plasma-concentration profiles for AOM[6] and TV-46000[7]
- D2RO and D2R antagonism were derived using established plasma concentration-response relationships[7,8]
- The models for estimating combined D2RO and antagonism were based on the principle of competitive binding, where both treatments compete for the same receptor site; occupancy is dictated by their concentrations and affinities
- Simulations modeled switching from AOM 400 mg (the most commonly used dose) to TV-46000 125 mg q1m or 250 mg q2m, 4 weeks (28 days) after the last AOM dose
- Simulations were performed to estimate D2RO and antagonism over time in virtual populations of 500 patients per treatment. Characteristics of the virtual cohorts were derived based on clinical trial populations

Switching to TV-46000 125 mg q1m or 250 mg q2m 4 weeks after the last dose of AOM 400 mg was estimated to preserve total D2R antagonism

- AOM 400 mg mean D2RO over 1 month was 90.9% at steady state, with 68.2% D2R antagonism before switching to TV-46000 125 mg q1m or TV-46000 250 mg q2m
- After the first TV-46000 q1m dose, mean D2RO was 71.2% for AOM and 19.2% for TV-46000 for a total of 90.4% receptor occupancy (**Figure 1**)
 - D2R antagonism was 53.4% for AOM and 19.2% for TV-46000 with a total antagonism of 72.6%
- After the first TV-46000 q2m dose, AOM mean D2RO was 62.5% and TV-46000 mean D2RO over 2 months was 26.7% for a total D2RO of 89.2% (**Figure 2**)
 - D2R antagonism was 46.8% for AOM and 26.7% for TV-46000 for a total antagonism of 73.6%
- As TV-46000 approached 70% to 80% of steady state (after 2 doses), D2R antagonism contribution of TV-46000 q1m increased to 35.9% and TV-46000 q2m to 50.0%, with AOM contributing 26.5% to 39.9% (**Table 1** and **Table 2**)
 - AOM's contribution to D2RO and D2R antagonism was <6% after 9 doses of TV-46000 q1m and 5 doses of TV-46000 q2m

Figure 1. Switching from AOM 400 mg to TV-46000 125 mg q1m

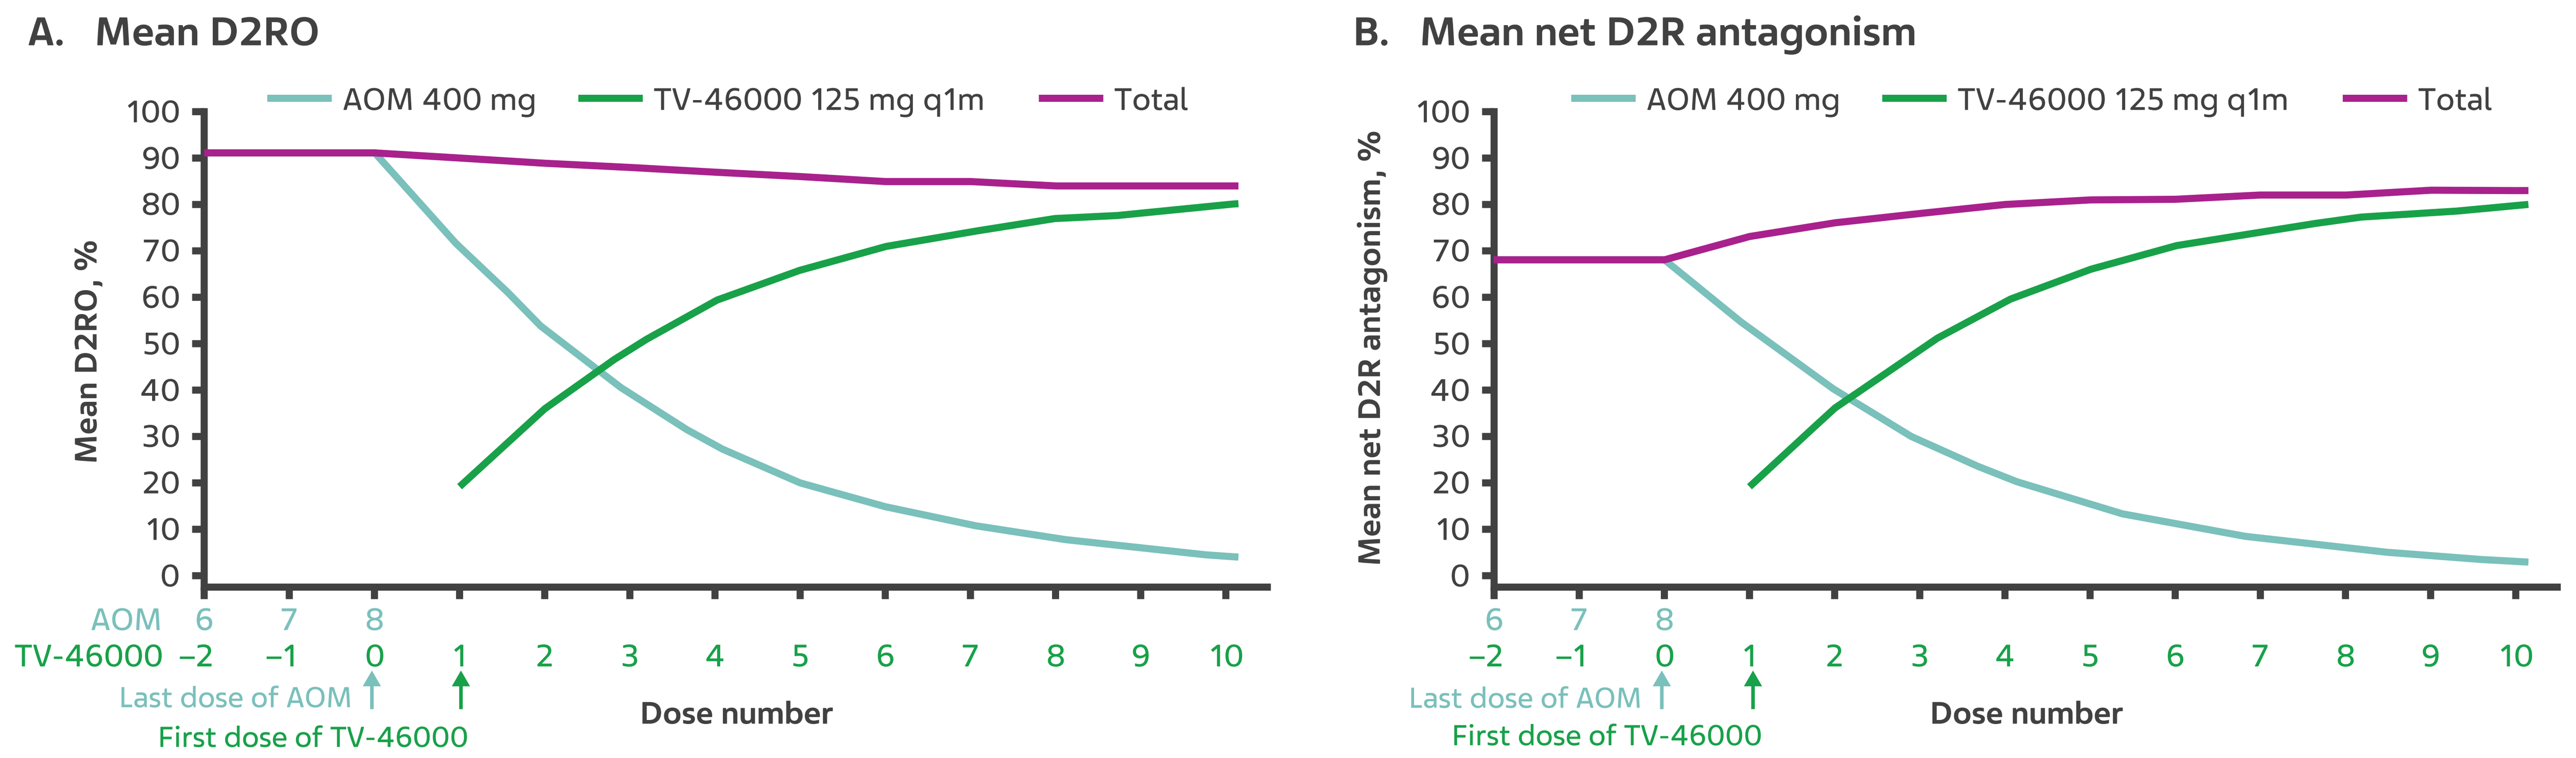
A. Mean D2RO
B. Mean net D2R antagonism
AOM 400 mg
TV-46000 125 mg q1m
Total
Mean D2RO, %
Mean net D2R antagonism, %
AOM 6 7 8
TV-46000 −2 −1 0 1 2 3 4 5 6 7 8 9 10
Last dose of AOM
First dose of TV-46000
Dose number

Table 1. Mean D2RO and D2R Antagonism for TV-46000 125 mg q1m and AOM 400 mg by Number of TV-46000 Doses

Number of TV-46000 doses after last dose of AOM	AOM 400 mg		TV-46000 125 mg q1m		Total	
	D2RO, %[a]	D2R antagonism, %	D2RO, %	D2R antagonism, %	D2RO, %	D2R antagonism, %
0 (Last dose of AOM)	90.9	68.2	0.0	0.0	90.9	68.2
1	71.2	53.4	19.2	19.2	90.4	72.6
2	53.2	39.9	35.9	35.9	89.1	75.8
3	39.0	29.2	48.9	48.9	87.8	78.1
4	28.2	21.1	58.5	58.5	86.7	79.7
5	20.3	15.2	65.5	65.5	85.8	80.8
10	4.2	3.2	79.6	79.6	83.8	82.8

[a]All percentages are presented as means over the dosing interval

Figure 2. Switching From AOM 400 mg to TV-46000 250 mg q2m

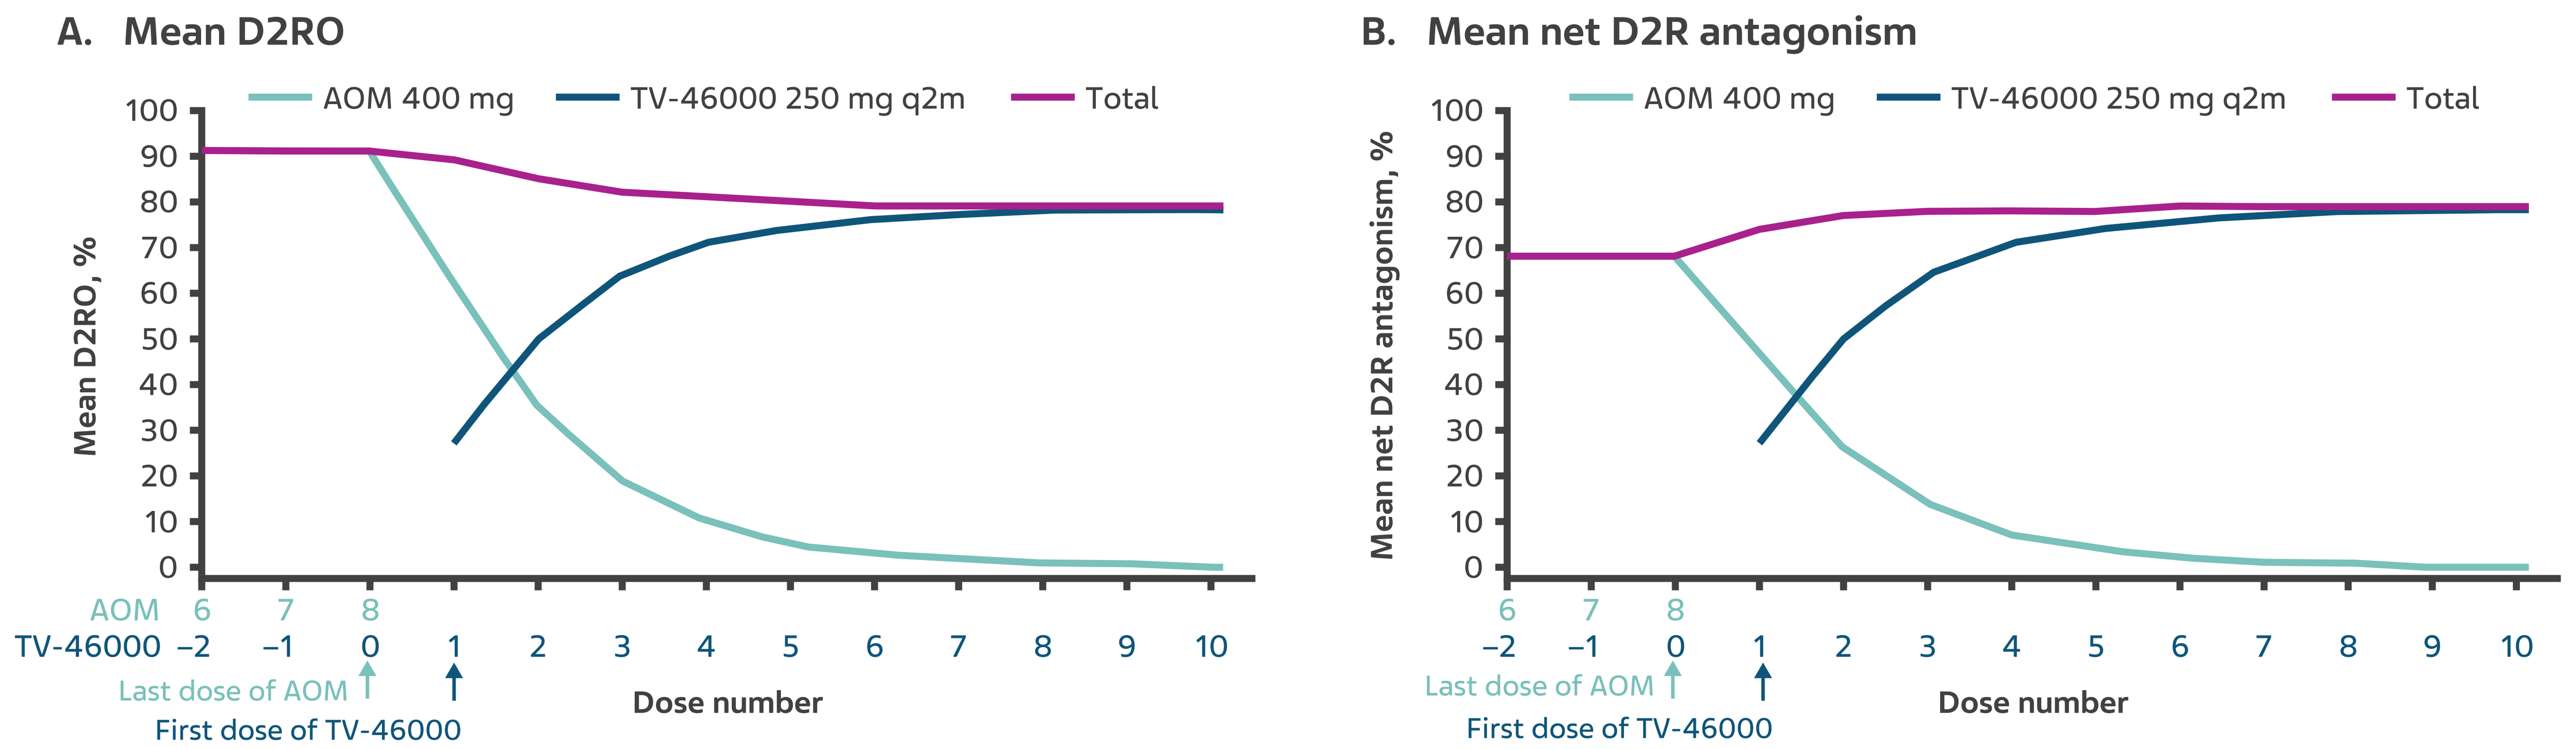
A. Mean D2RO
B. Mean net D2R antagonism
AOM 400 mg
TV-46000 250 mg q2m
Total
Mean D2RO, %
Mean net D2R antagonism, %
AOM 6 7 8
TV-46000 −2 −1 0 1 2 3 4 5 6 7 8 9 10
Last dose of AOM
First dose of TV-46000
Dose number

Table 2. Mean D2RO and D2R Antagonism for TV-46000 250 mg q2m and AOM 400 mg by Number of TV-46000 Doses

Number of TV-46000 doses after last dose of AOM	AOM 400 mg		TV-46000 250 mg q2m		Total	
	D2RO, %[a]	D2R antagonism, %	D2RO, %	D2R antagonism, %	D2RO, %	D2R antagonism, %
0 (Last dose of AOM)	90.9	68.2	0.0	0.0	90.9	68.2
1	62.5	46.8	26.7	26.7	89.2	73.6
2	35.3	26.5	50.0	50.0	85.3	76.5
3	18.8	14.1	63.7	63.7	82.5	77.8
4	9.9	7.5	70.8	70.8	80.7	78.3
5	5.4	4.1	74.4	74.4	79.8	78.4
10	0.4	0.3	78.3	78.3	78.8	78.7

[a]All percentages are presented as means over the dosing interval

Conclusions

- Simulations estimated that switching from AOM 400 mg to TV-46000 125 mg q1m or 250 mg q2m resulted in numerically higher total net D2R antagonism over the 10 doses simulated
- These simulations demonstrate the relative differences in postsynaptic activity after switching from relevant doses of a D2R partial agonist antipsychotic (AOM 400 mg) to a D2R antagonist TV-46000 125 mg q1m or 250 mg q2m
- These findings are limited as the simulations do not evaluate the pharmacodynamic effects on other receptors and results are not based on direct clinical data
- Clinical decisions should be guided by clinician judgement, patient preference, convenience, and tolerability

Acknowledgments

Medical writing and editorial support were provided by Jean-Paul Fouche, PhD, Alison Adams, PhD, CMPP, and Kelsey Gribbon, MS, of Ashfield MedComms, an Inizio company, and were funded by Teva Branded Pharmaceutical Products R&D LLC.

Disclosures

This study was supported by Teva Branded Pharmaceutical Products R&D LLC. Itay Perlstein has received consultancy fees from Teva Pharmaceuticals in relation to this study. Jonathan Meyer has received consulting fees or honoraria from Teva Pharmaceuticals. Sharath Kumar, Vijay Ivaturi, Nikita Ramwani, and Pavan Rathina Muthu Kumar are employees of Pumas-AI, which has received payments from Teva Pharmaceuticals in relation to this study. Kelli R. Franzenburg, Mark Suett, Rolf Hansen, Avia Merenlender Wagner, Arti Phatak, and Rajendra Singh are employees and shareholders of Teva Pharmaceuticals.

Abbreviations

AOM = aripiprazole monohydrate once monthly, D2R = D2 receptor, D2RO = D2-receptor occupancy, LAI = long-acting injectable antipsychotic, q1m = once monthly, q2m = once every 2 months

References

1. Højlund M, et al. *Expert Opin Pharmacother*. 2023;24:1463–1489.
2. Uzedy. Prescribing information. Teva Neuroscience, Inc; 2025.
3. Kim E, et al. *J Cereb Blood Flow Metab*. 2011;32:759–768.
4. Olsen CK, et al. *Eur J Pharmacol*. 2008;584:318–327.
5. McCutcheon RA, et al. *World Psychiatry*. 2020;19:15–33.
6. Wang X, et al. *Clin Pharmacol Drug Dev*. 2022;11:150–164.
7. Perlstein I, et al. *Neurol Ther*. 2025;14:829–848.
8. Raoufinia A, et al. *Int J Neuropsychopharmacol*. 2017;20:295–304.

Presented at the Annual Psych Congress; September 17–21, 2025; San Diego, CA.